Abstract #4363

A First-in-Man Phase 1 Study Of CUDC-907, a First-in-Class Chemically-Designed Dual Inhibitor of PI3K and HDAC in Patients With Refractory or Relapsed Lymphoma and Multiple Myeloma

Anas Younes, MD[1], Ian W. Flinn, MD, PhD[2], Yasuhiro Oki, MD[3], Amanda Copeland, RN, MSN, CNS[4], Ali Fattaey, PhD[5], Cheng-Jung Lai, PhD[5], Robert Laliberte, MS[5], Maurizio Voi, MD[5] and Jesus G Berdeja, MD[2]

[1]Department of Medicine, Memorial Sloan-Kettering Cancer Center, New York, NY; [2]Sarah Cannon Cancer Institute, Nashville, TN; [3]Lymphoma and Myeloma Dept, M.D. Anderson Cancer Center, Houston, TX; [4]Memorial Sloan-Kettering Cancer Center, New York, NY; [5]Curis, Inc., Lexington, MA

Introduction

Histone deacetylases (HDACs) remove acetyl groups from histone and non-histone proteins, thereby playing an important role in the regulation of oncogenic gene expression and protein activity. As a consequence, HDAC inhibition induces multi-node epigenetic modifications of cancer signaling networks. The HDAC inhibitors vorinostat and romidepsin have been approved by the U.S. Food and Drug Administration for the treatment of cutaneous T-cell lymphoma and peripheral T-cell lymphoma. In addition, clinical activity with HDAC inhibitors has also been observed in Hodgkin's lymphoma, diffuse large B-cell lymphoma (DLBCL), multiple myeloma (MM), and acute myeloid leukemia (AML).

The Class I phosphoinositide 3-kinase (PI3K) family of enzymes consists of four closely related isoforms (p110α, p110β, p110δ and p110γ) that generate phospholipid second messengers and integrate signals from multiple receptor tyrosine kinases to govern cell proliferation, survival, migration, proliferation, apoptosis, neovascularization, and metastasis. The potential oncogenicity of PI3-kinases was revealed by the occurrence of gain-of-function mutations in PIK3CA, the gene coding for the catalytic subunit p110α. While several orally active PI3K inhibitors are currently in clinical development and have demonstrated activity in a variety of hematological malignancies, none has received regulatory approval to date.

CUDC-907 is a small molecule that combines the active hydroxamate moiety of HDAC inhibitors with a PI3K inhibitor morpholinopyrimidine pharmacophore. CUDC-907 potently inhibits class I PI3K (alpha, beta, and delta) as well as HDAC class I and II enzymes. Preclinical experiments demonstrated that CUDC-907 inhibits the PI3K-AKT-mTOR pathway and compensatory MEK/ERK and STAT3 signaling pathways. CUDC-907 shows greater growth inhibition and proapoptotic activity than single-target PI3K or HDAC inhibitors in both cultured and implanted cancer cells, including human B-cell tumor xenograft models.

Here we present the preliminary results of a First-in-Man Phase 1 Study of CUDC-907.

CUDC-907 Target Activity

HDAC	IC50 (nM)
1	1.7
2	5.0
3	1.8
6	27
10	2.8
4, 5, 7, 8, 9	>200

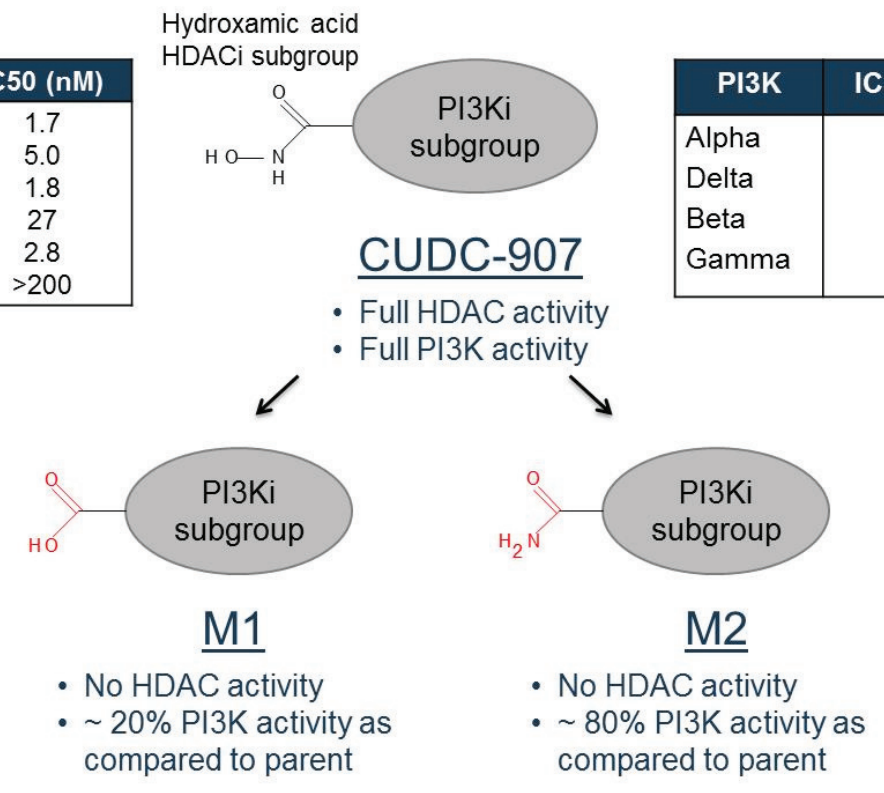

Hydroxamic acid HDACi subgroup
PI3Ki subgroup
CUDC-907
• Full HDAC activity
• Full PI3K activity
PI3Ki subgroup
PI3Ki subgroup
M1
• No HDAC activity
• ~ 20% PI3K activity as compared to parent
M2
• No HDAC activity
• ~ 80% PI3K activity as compared to parent

PI3K	IC50 (nM)
Alpha	19
Delta	39
Beta	54
Gamma	311

CONFLICT OF INTEREST DISCLOSURE:

No relevant conflicts of interest to disclose: AY, IWF, YO, AC, JGB; Curis employees: AF, CJL, RL, MV.

Partial funding for this study is being provided by The Leukemia & Lymphoma Society.

Study Objectives & Design

Primary:

- To determine the maximum tolerated dose (MTD) and recommended Phase 2 dose (RP2D) of oral CUDC-907 in patients with relapsed or refractory lymphoma or MM

Secondary:

- To assess the safety and tolerability of CUDC-907
- To assess the pharmacokinetics (PK) of CUDC-907
- To evaluate biomarkers of CUDC-907 activity
- To assess preliminary anti-cancer activity of CUDC-907

Overall Design

- Open-label, multicenter, nonrandomized Phase I study
- Standard 3+3 dose escalation design of CUDC-907 administered once daily (QD); intermittent dosing schedules were added by protocol amendment, including once daily twice weekly (BIW) or once daily thrice weekly (TIW)
- Subjects will be enrolled in consecutive cohorts at dose levels of 30 mg, 60 mg, 90 mg, 120 mg, 150 mg or 180 mg/day
- Dose limiting toxicity (DLT) is defined as any of the following adverse events (AE) occurring up to 7 days following completion of study drug dosing, regardless of relationship to study drug unless clearly related to the underlying disease:
 - Non-hematological Grade 3 AE, other than Grade 3 nausea or vomiting in subjects treated with less than optimal antiemetic therapy
 - Any AE resulting in a dose delay ≥7 days
 - Grade 4 neutropenia lasting ≥7 days, or ≥Grade 3 with fever >101.3°F (38.5°C) or infection
 - Grade 4 thrombocytopenia ≥7 days, or ≥Grade 3 with significant bleeding

Study Population

- Subjects with histopathologically confirmed diagnosis of lymphoma or relapsed/refractory MM after ≥2 prior regimens
- Measurable or evaluable disease
- Age ≥ 18 years
- ECOG performance status ≤2
- Adequate bone marrow and organ function
- Prior treatment with a PI3K inhibitor allowed

Drug Administration

- CUDC-907 will be administered orally according to the assigned dose scheduled, with meals (±30 minutes), in 21 day cycles

PK/PD Evaluations

- PK assessments on Days 1 & 15 in Cycle 1
- PBMC and plasma biomarker samples on Cycle 1 Days 1, 8 & 15
- Optional tissue sampling (skin, tumor, hair follicle and/or bone marrow) within 7 days prior to initiating CUDC-907 dosing and 2-6 hours after CUDC-907 dose on Day 15 through Cycle 2 Day 1

Tumor Assessment

- Per Revised Response Criteria for Malignant Lymphoma or International Uniform Response Criteria for Multiple Myeloma

Patient Characteristics and Disposition

Parameter (n)	30 mg QD (N=7)	60 mg QD (N=3)	60 mg BIW (N=3)	Overall (N=13)
Male	5	2	2	9
Female	2	1	1	4
Age (mean), yrs	59.4	70.0	68.7	64.0
Histology				
Non-Hodgkin's Lymphoma				
Small lymphocytic	1	0	1	2
Diffuse large B-cell	0	2	0	2
Mantle cell	0	0	2	2
Lymphoplasmacytic	1	0	0	1
Follicular	1	0	0	1
Follicular/DLBCL	1	0	0	1
Hodgkin's Lymphoma	1	1	0	2
Multiple Myeloma	2	0	0	2
Stage at Study Entry				
I-II	2	0	0	2
III-IV	5	2	3	10
Unknown	0	1	0	1
Prognostic Score (IPI, FLIPI or MIPI)				
Low	1	0	0	1
Intermediate	2	3	3	8
High	2	0	0	2
Prior Treatments				
# prior regimens [median (range)]	5 (2-8)	3 (2-9)	3 (2-5)	4 (2-9)
Bone Marrow Transplant	2	1	1	4
No. Discontinued Study Treatment	4	2	1	7
Progressive disease	1	1	1	3
Subject/Physician decision	3	0	0	3
Treatment-related toxicity (DLT)	0	1	0	1

Adverse Events

Adverse Events Reported in ≥2 Subjects

	30 mg QD (N=7)		60 mg QD (N=3)		60 mg BIW (N=3)		Overall (N=13)
Maximum Grade:	**1-2**	**3-4**	**1-2**	**3-4**	**1-2**	**3-4**	**N (%)**
Any (Maximum severity)	4	3	0	3	1	0	11 (84.6)
Dose Limiting Toxicity	0	0	0	1	0	0	1 (7.6)
Diarrhea	7		2	1	0		10 (76.9)
Thrombocytopenia/decr. platelet	2	3	1	2	0		8 (61.5)
Fatigue	6		1		1		8 (61.5)
Nausea	3		1		0		4 (30.8)
Neutropenia/decr. ANC	1	1	1		0		3 (23.1)
ALP increased	2		0		0		2 (15.4)
Appetite decreased	2		0		0		2 (15.4)
Hypotension	2		0		0		2 (15.4)
Pyrexia	2		0		0		2 (15.4)

- 2 DLTs (Grade 3 diarrhea & Grade 4 hyperglycemia) were reported for 1 subject at the 60 mg QD dose level
- 1 subject at 60 mg QD was dose reduced to 30 mg QD during Cycle 1 due to Grade 2 diarrhea
- 2 subjects experienced treatment-related SAEs (Grade 3 epistaxis at 30 mg QD, Grade 3 diarrhea & Grade 4 hyperglycemia at 60 mg QD)
- MTD for the QD schedule was determined to be 30 mg, due to treatment-related AEs & dosing interruptions for neutropenia and/or thrombocytopenia

PK/PD Results

Clinical Plasma PK (Mean)

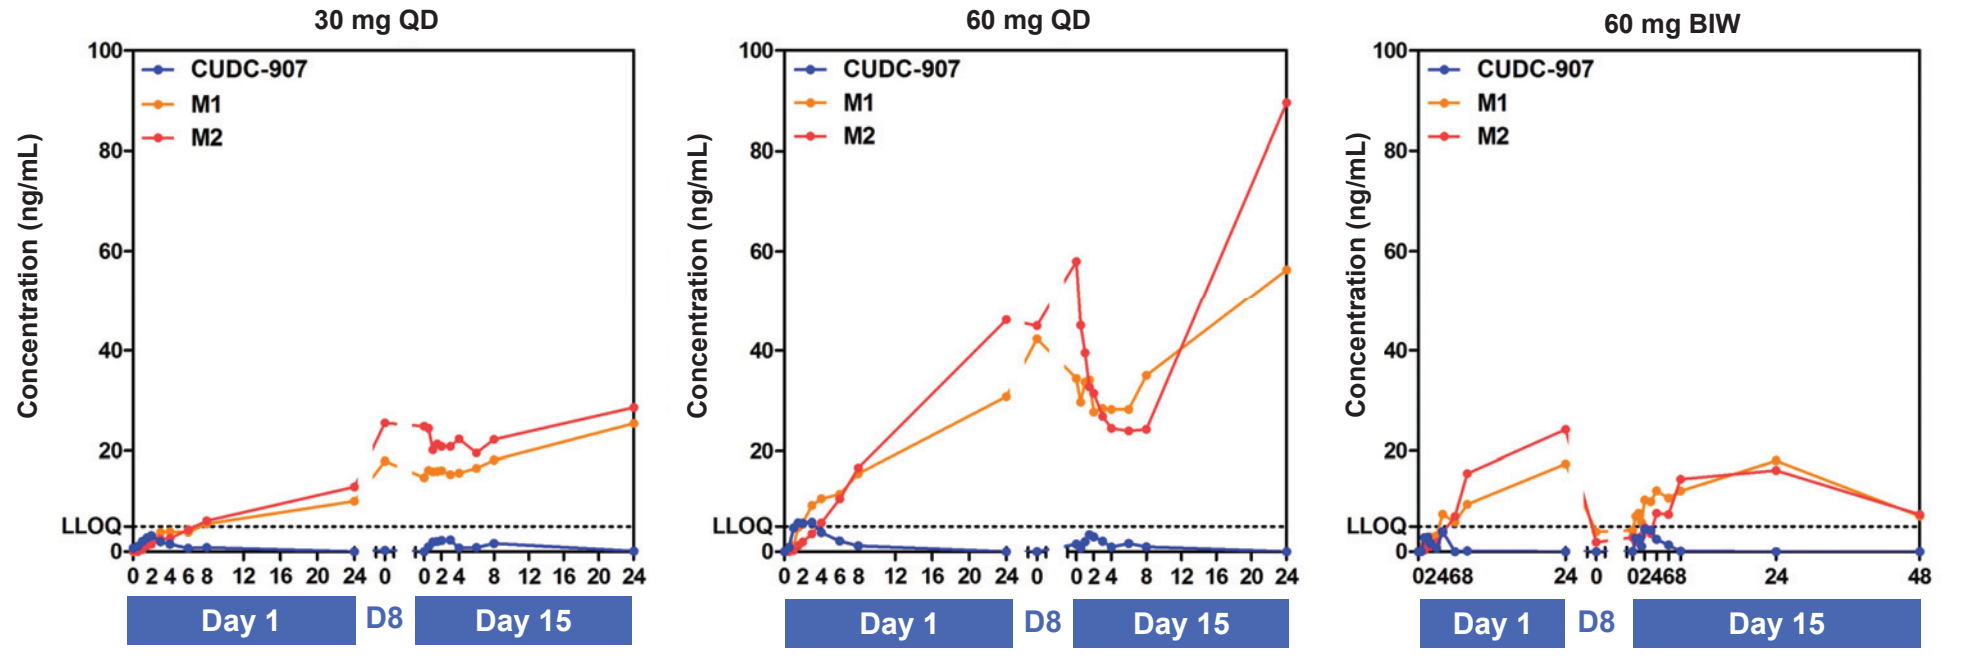

30 mg QD
60 mg QD
60 mg BIW
CUDC-907
M1
M2
Concentration (ng/mL)
LLOQ
Day 1
D8
Day 15

- Clinical plasma levels show low CUDC-907 exposure and accumulation of M1 and M2 with QD dosing
- Analysis of PBMC, plasma and tissue samples for PD analyses is planned/ongoing
- Clinical PK/PD observations may be informed by the following mouse tumor model studies:

CUDC-907 & M2 Distribution in Mouse Model (25 mg/kg PO QD)

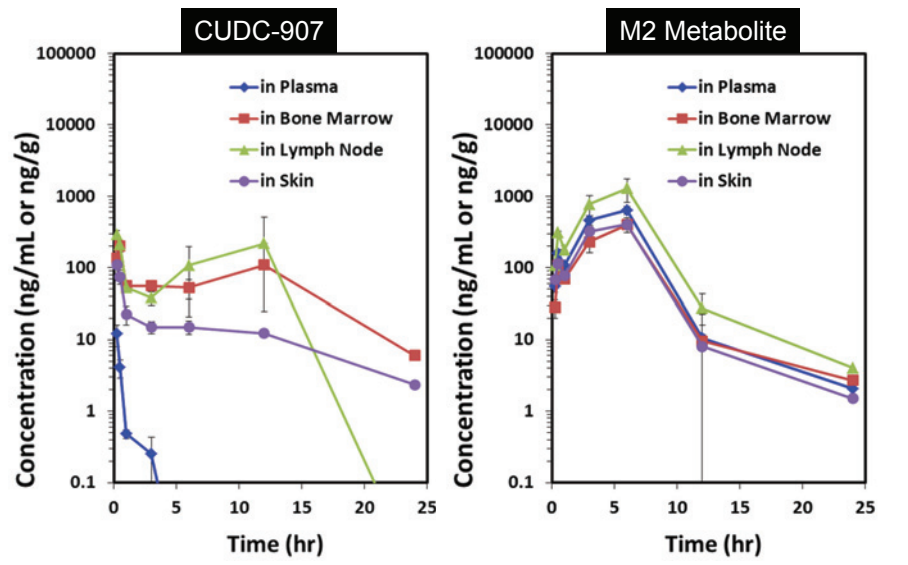

CUDC-907
M2 Metabolite
in Plasma
in Bone Marrow
in Lymph Node
in Skin
Concentration (ng/mL or ng/g)
Time (hr)

Efficacy and Biomarker PD Effect in Mouse Model

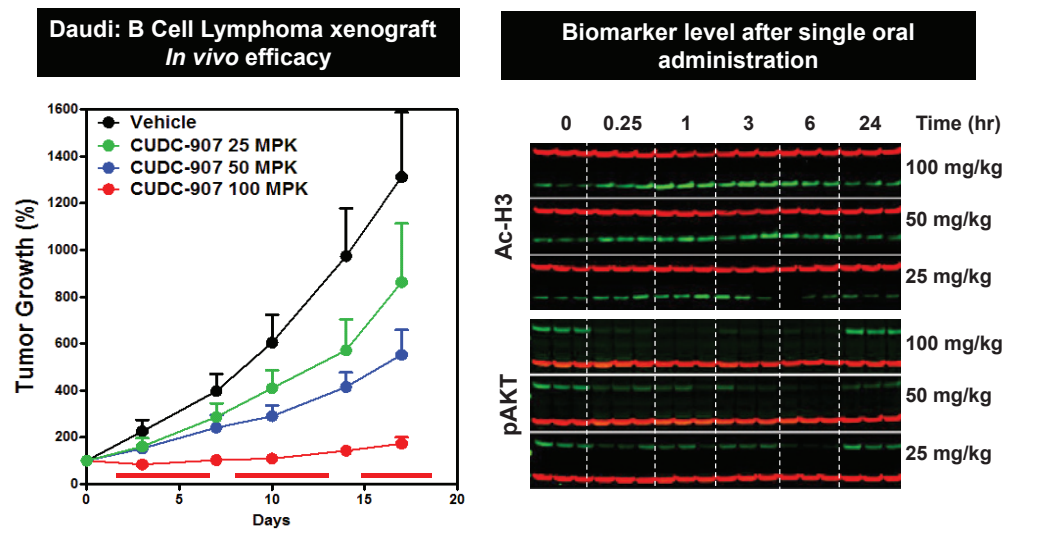

Daudi: B Cell Lymphoma xenograft
In vivo efficacy
Vehicle
CUDC-907 25 MPK
CUDC-907 50 MPK
CUDC-907 100 MPK
Tumor Growth (%)
Days
Biomarker level after single oral administration
0
0.25
1
3
6
24
Time (hr)
Ac-H3
pAKT
100 mg/kg
50 mg/kg
25 mg/kg
100 mg/kg
50 mg/kg
25 mg/kg

- In the mouse tumor model:
 - CUDC-907 is predominantly distributed to tissue, with low plasma exposure
 - No M1 and M2 accumulation with QD dosing
 - PD effects in tumor tissue were demonstrated as a rapid increase in acetylated histone H3 (Ac-H3) (HDAC inhibition) & sustained reduction in phospho-AKT levels (PI3K inhibition)

Best Tumor Response

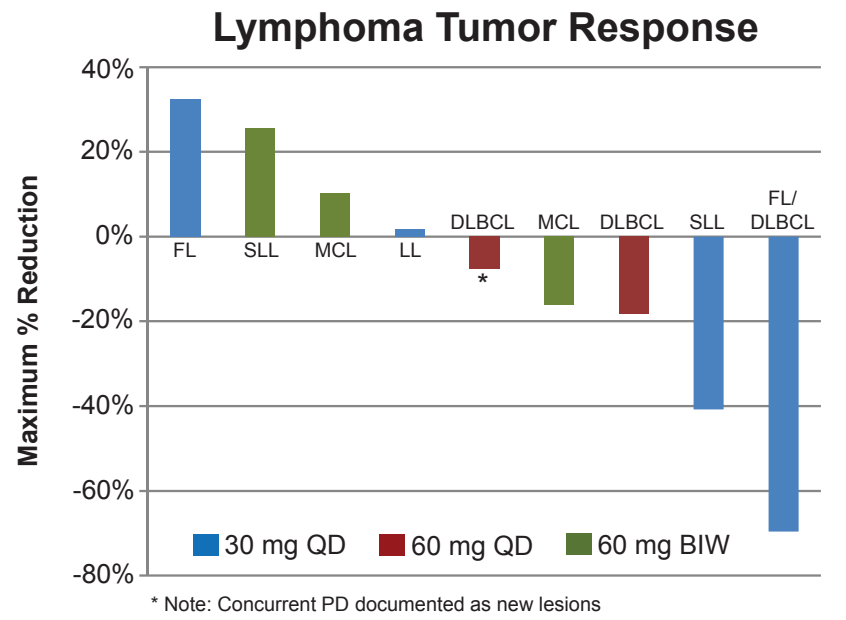

Lymphoma Tumor Response
Maximum % Reduction
FL
SLL
MCL
LL
DLBCL
MCL
DLBCL
SLL
FL/DLBCL
30 mg QD
60 mg QD
60 mg BIW
* Note: Concurrent PD documented as new lesions

- 11 subjects have at least 1 post-treatment response assessment and are evaluable for efficacy analysis
- 1 subject with mixed FL/DLBCL enrolled at the 30 mg QD dose level achieved a partial response (70% reduction in a single target lesion) in Cycle 4
- 7 subjects have stable disease (SD) as best response, including 4 with SD ≥4 cycles of study treatment
- 1 subject with MM is currently in Cycle 13 of study treatment with SD

Conclusions

- QD dosing with CUDC-907 was associated with diarrhea, thrombocytopenia and fatigue, limiting the ability to dose escalate using this schedule
- Intermittent dosing appears to be better tolerated. Dose escalation continues into BIW and TIW cohorts.
- Low CUDC-907 plasma exposure at dose levels studied thus far is consistent with non-clinical studies that demonstrated far greater exposure in tissue as compared to plasma
- Preliminary evidence of anti-tumor activity based on 1 objective response (N=1, PR in FL/DLBCL)

55th ASH Annual Meeting and Exposition, 7-10 December 2013 New Orleans, LA